An *in silico* structural approach to critical quality attributes assessment of biopharmaceutical products

Saporiti S[1,2], Parravicini C[1], Eberini I[2], Centola F[1], Pergola C[1] and Sensi C[1]

[1] Analytical System Center of Expertise, Merck Serono, Guidonia Montecelio-Rome, Italy
[2] Department of Pharmacological and Biomolecular Sciences, University of Milan, Italy

BACKGROUND

Currently, monoclonal antibodies (mAbs) are one of the most innovative class of biopharmaceuticals in development, due their ability to specifically recognize molecular targets.
Pharmaceutical companies have implemented a new strategy to control the quality profile of mAbs during the development phase: the "Quality by Design" (Qbd) approach. QbD is essentially focused on critical quality attributes (CQAs) assessment. CQAs are defined as all the physical, chemical, biological or microbiological properties that should be within an appropriate limit, range, or distribution to ensure the desired product quality[1]. These attributes have an impact on bioactivity, PK, immunogenicity and safety and are generally associated with the drug substance, excipients, intermediates and pharmaceutical product. In this global approach, the introduction of a structural investigation, can be very useful to early identify and assess potential CQAs (pCQAs) and for drug substance and excipients understanding.

AIMS

- Structural data collection about overall IgG1
- Identification of suitable templates
- Homology modeling of both IgG1 κ and λ and structural analysis of whole mAbs
- Identification of differences between κ and λ chains by bioinformatics tools

IgG1: STRUCTURAL INSIGHTS

IgG1s are the most expressed immunoglobulin sub-class in humans. They have a "Y-shaped" conformation, due their structural organization: two heterodimers of heavy and light chains (HC and LC), linked each other by interchain disulphide bonds, assembly to form a Y-architecture.

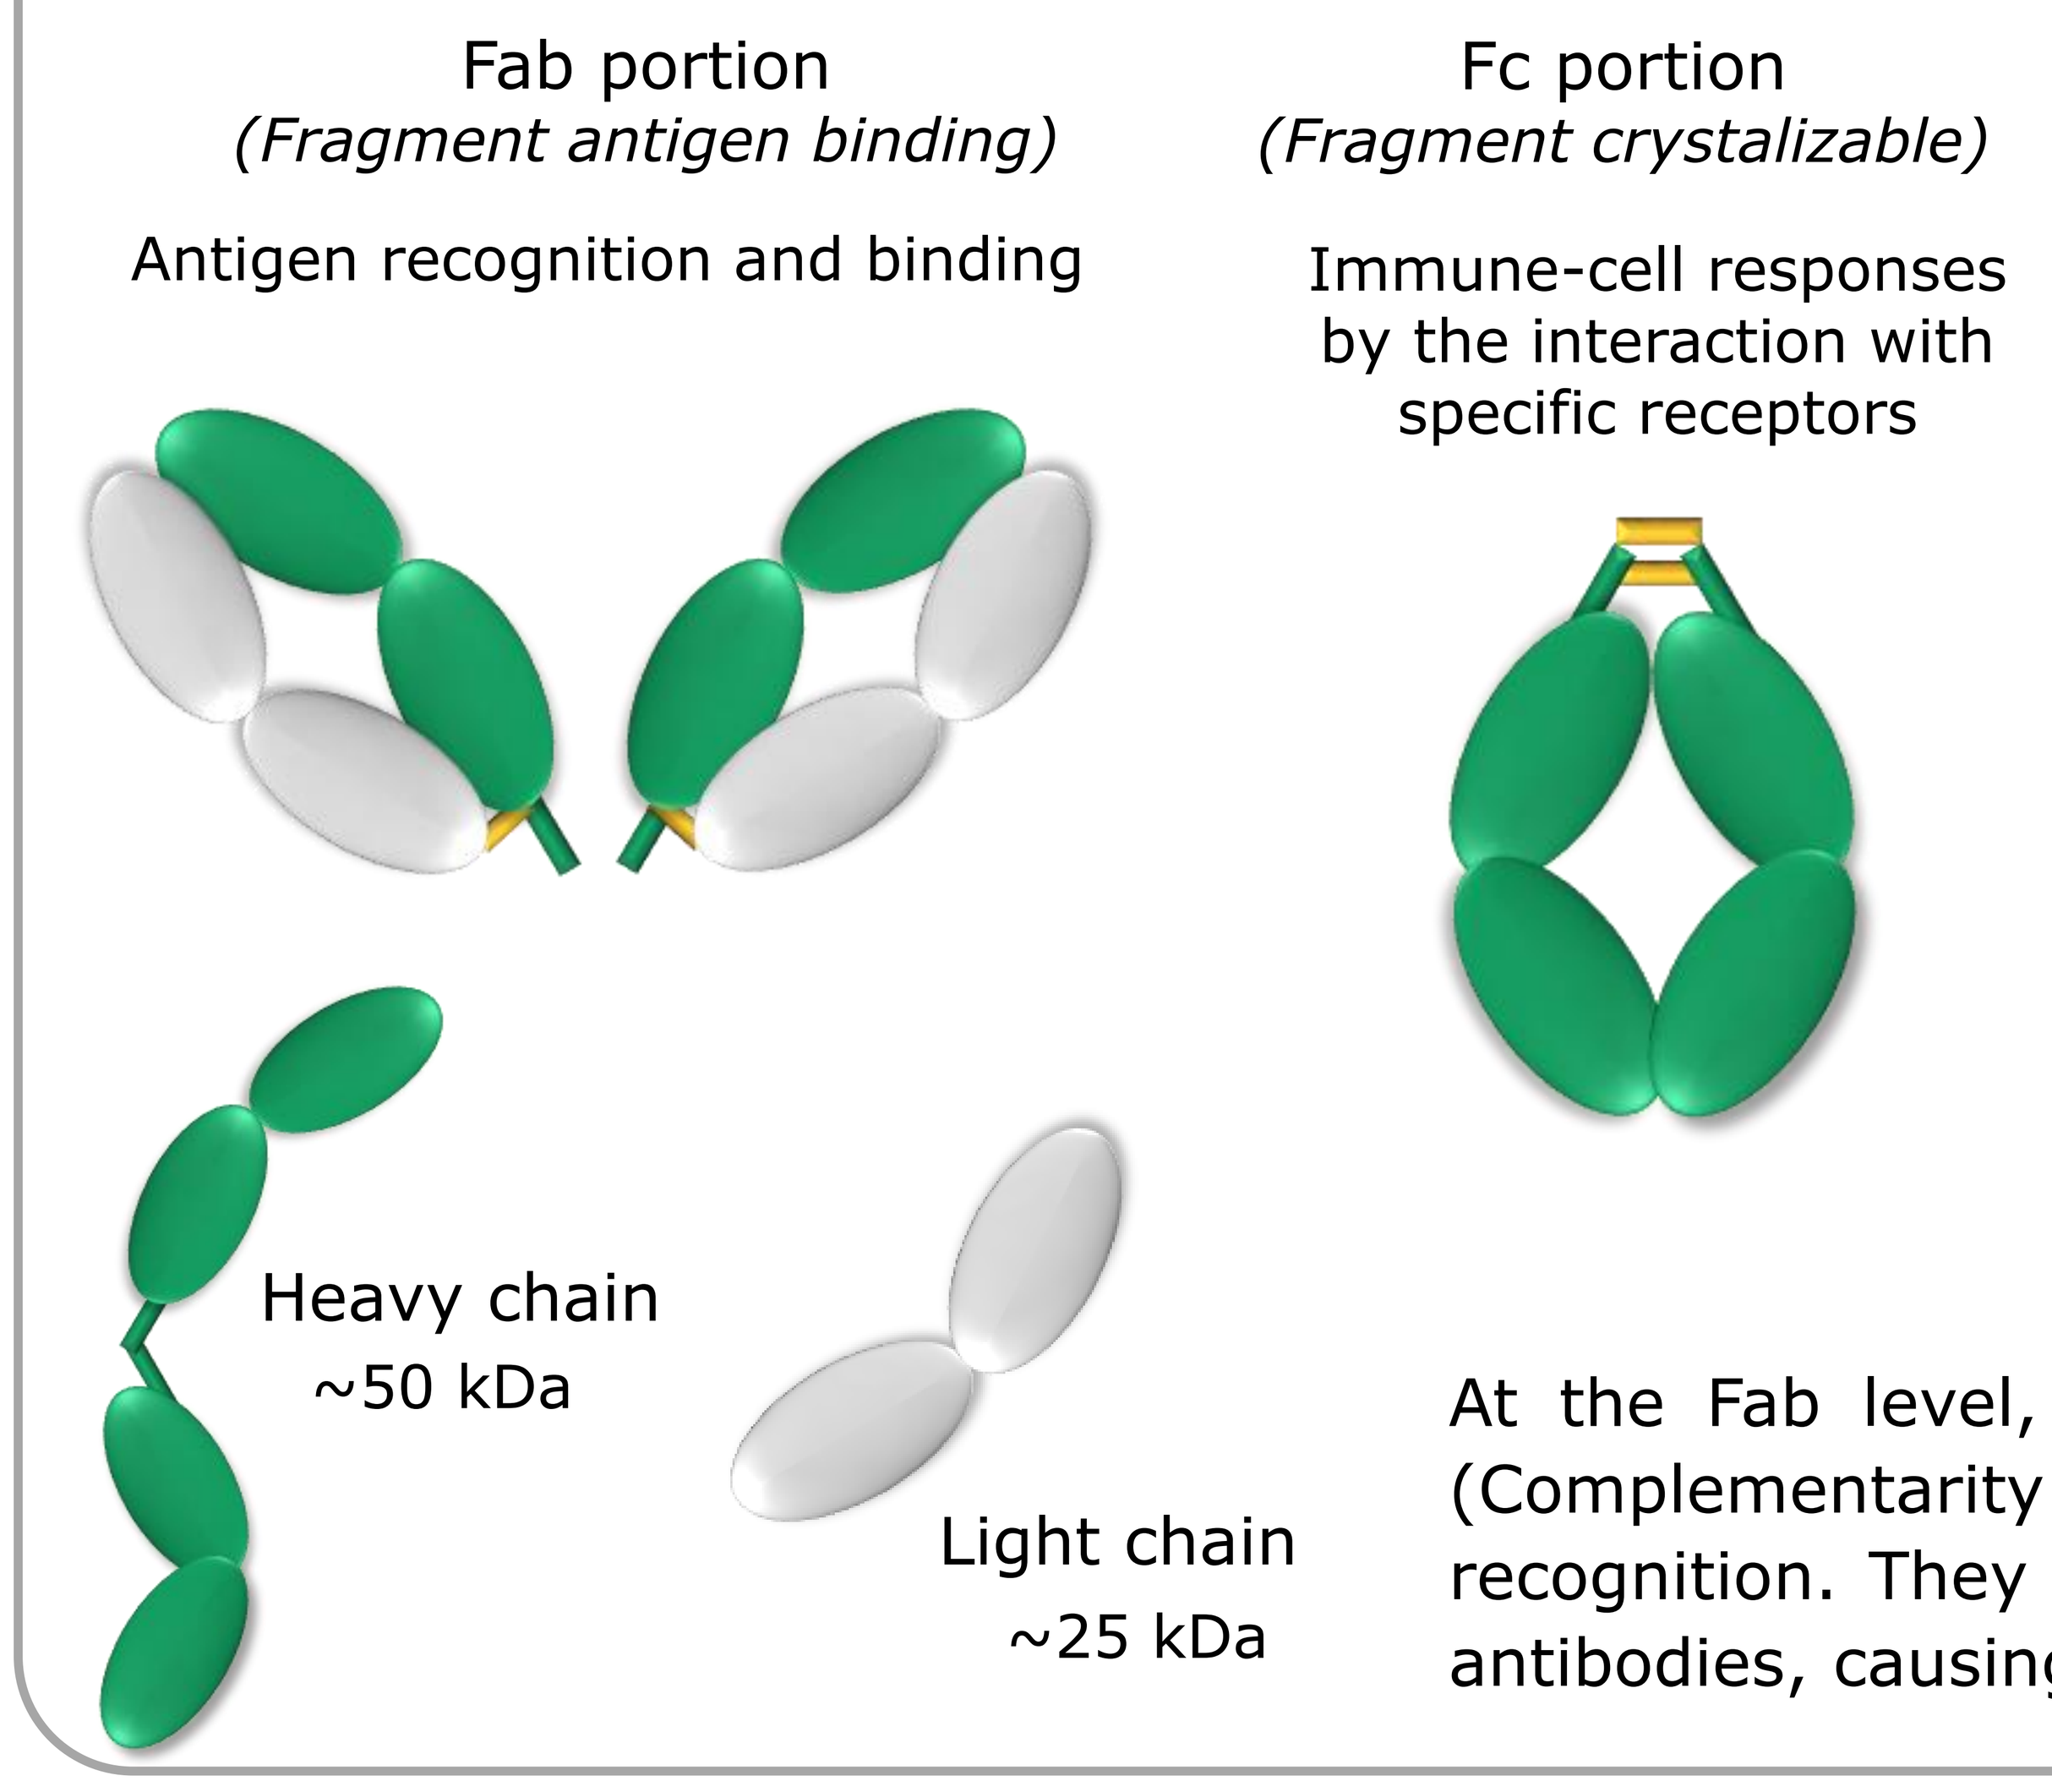
Fab portion
(Fragment antigen binding)
Antigen recognition and binding
Fc portion
(Fragment crystalizable)
Immune-cell responses by the interaction with specific receptors
Heavy chain
~50 kDa
Light chain
~25 kDa

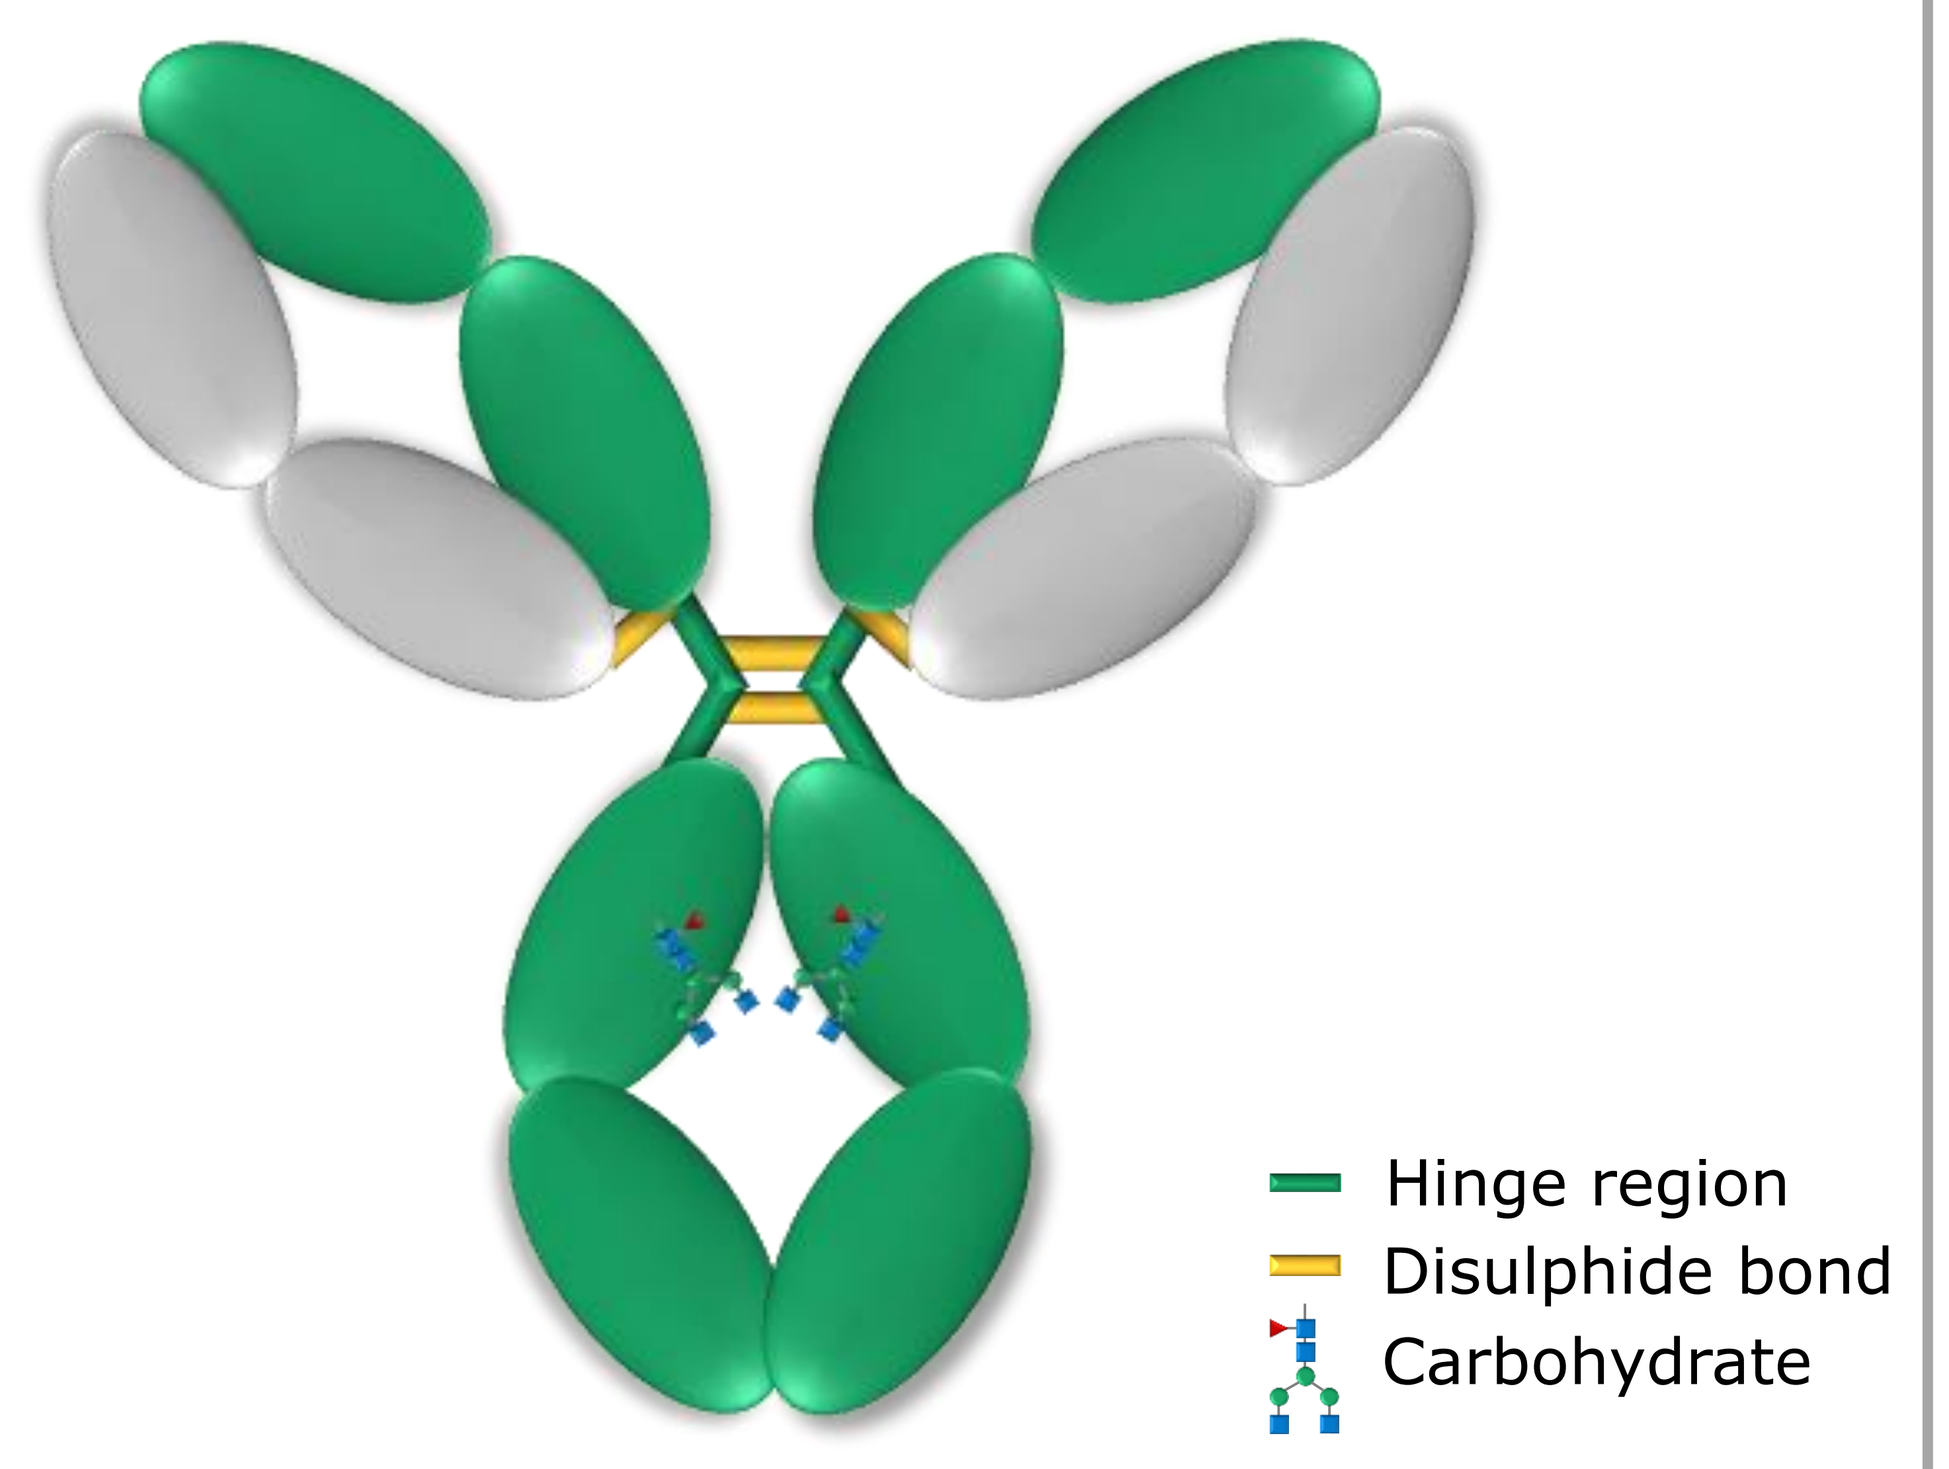
Hinge region
Disulphide bond
Carbohydrate

At the Fab level, the three hypervariability regions, called CDRs (Complementarity Determining Regions), drive the antigen recognition. They differ in sequence and in length among different antibodies, causing the specificity and affinity for the target[2].

IgG1λ and IgG1κ MODELING

Due to the lack of experimental data on complete IgG1s structures, we developed an *in silico* strategy to predict the full structure of this antibodies class. We focused our study on the two subtypes of light chain, λ and κ, due their different physical-chemical properties[3,4] and the impact of this feature on molecules bioactivity. Usually, for therapeutic mAbs, only the X-ray structure of Fab portions is solved. So, we used a homology modeling chimeric approach to obtain a prediction of whole 3D structure of proteins by a combination of different templates.
In detail, we investigated the atomistic structure of two therapeutic and commercially available mAbs: avelumab, a recently approved anti-PDL1 IgG1λ, indicated for Merkell-cell carcinoma[5], and adalimumab, an anti-TNFα IgG1κ used in the treatment of rheumatoid and psoriatic arthritis and for the Crohn disease[6]. We decided to use as template to model the Fab portion 4NKI.pdb[7] and 4NYL.pdb, respectively, the X-ray structures of avelumab and adalimumab Fab. Then, we used 1HZH.pdb[8], the only one human fully crystalized IgG1, as Fc and backbone template for both mAbs. All the computational procedures were carried out by MOE software[9].

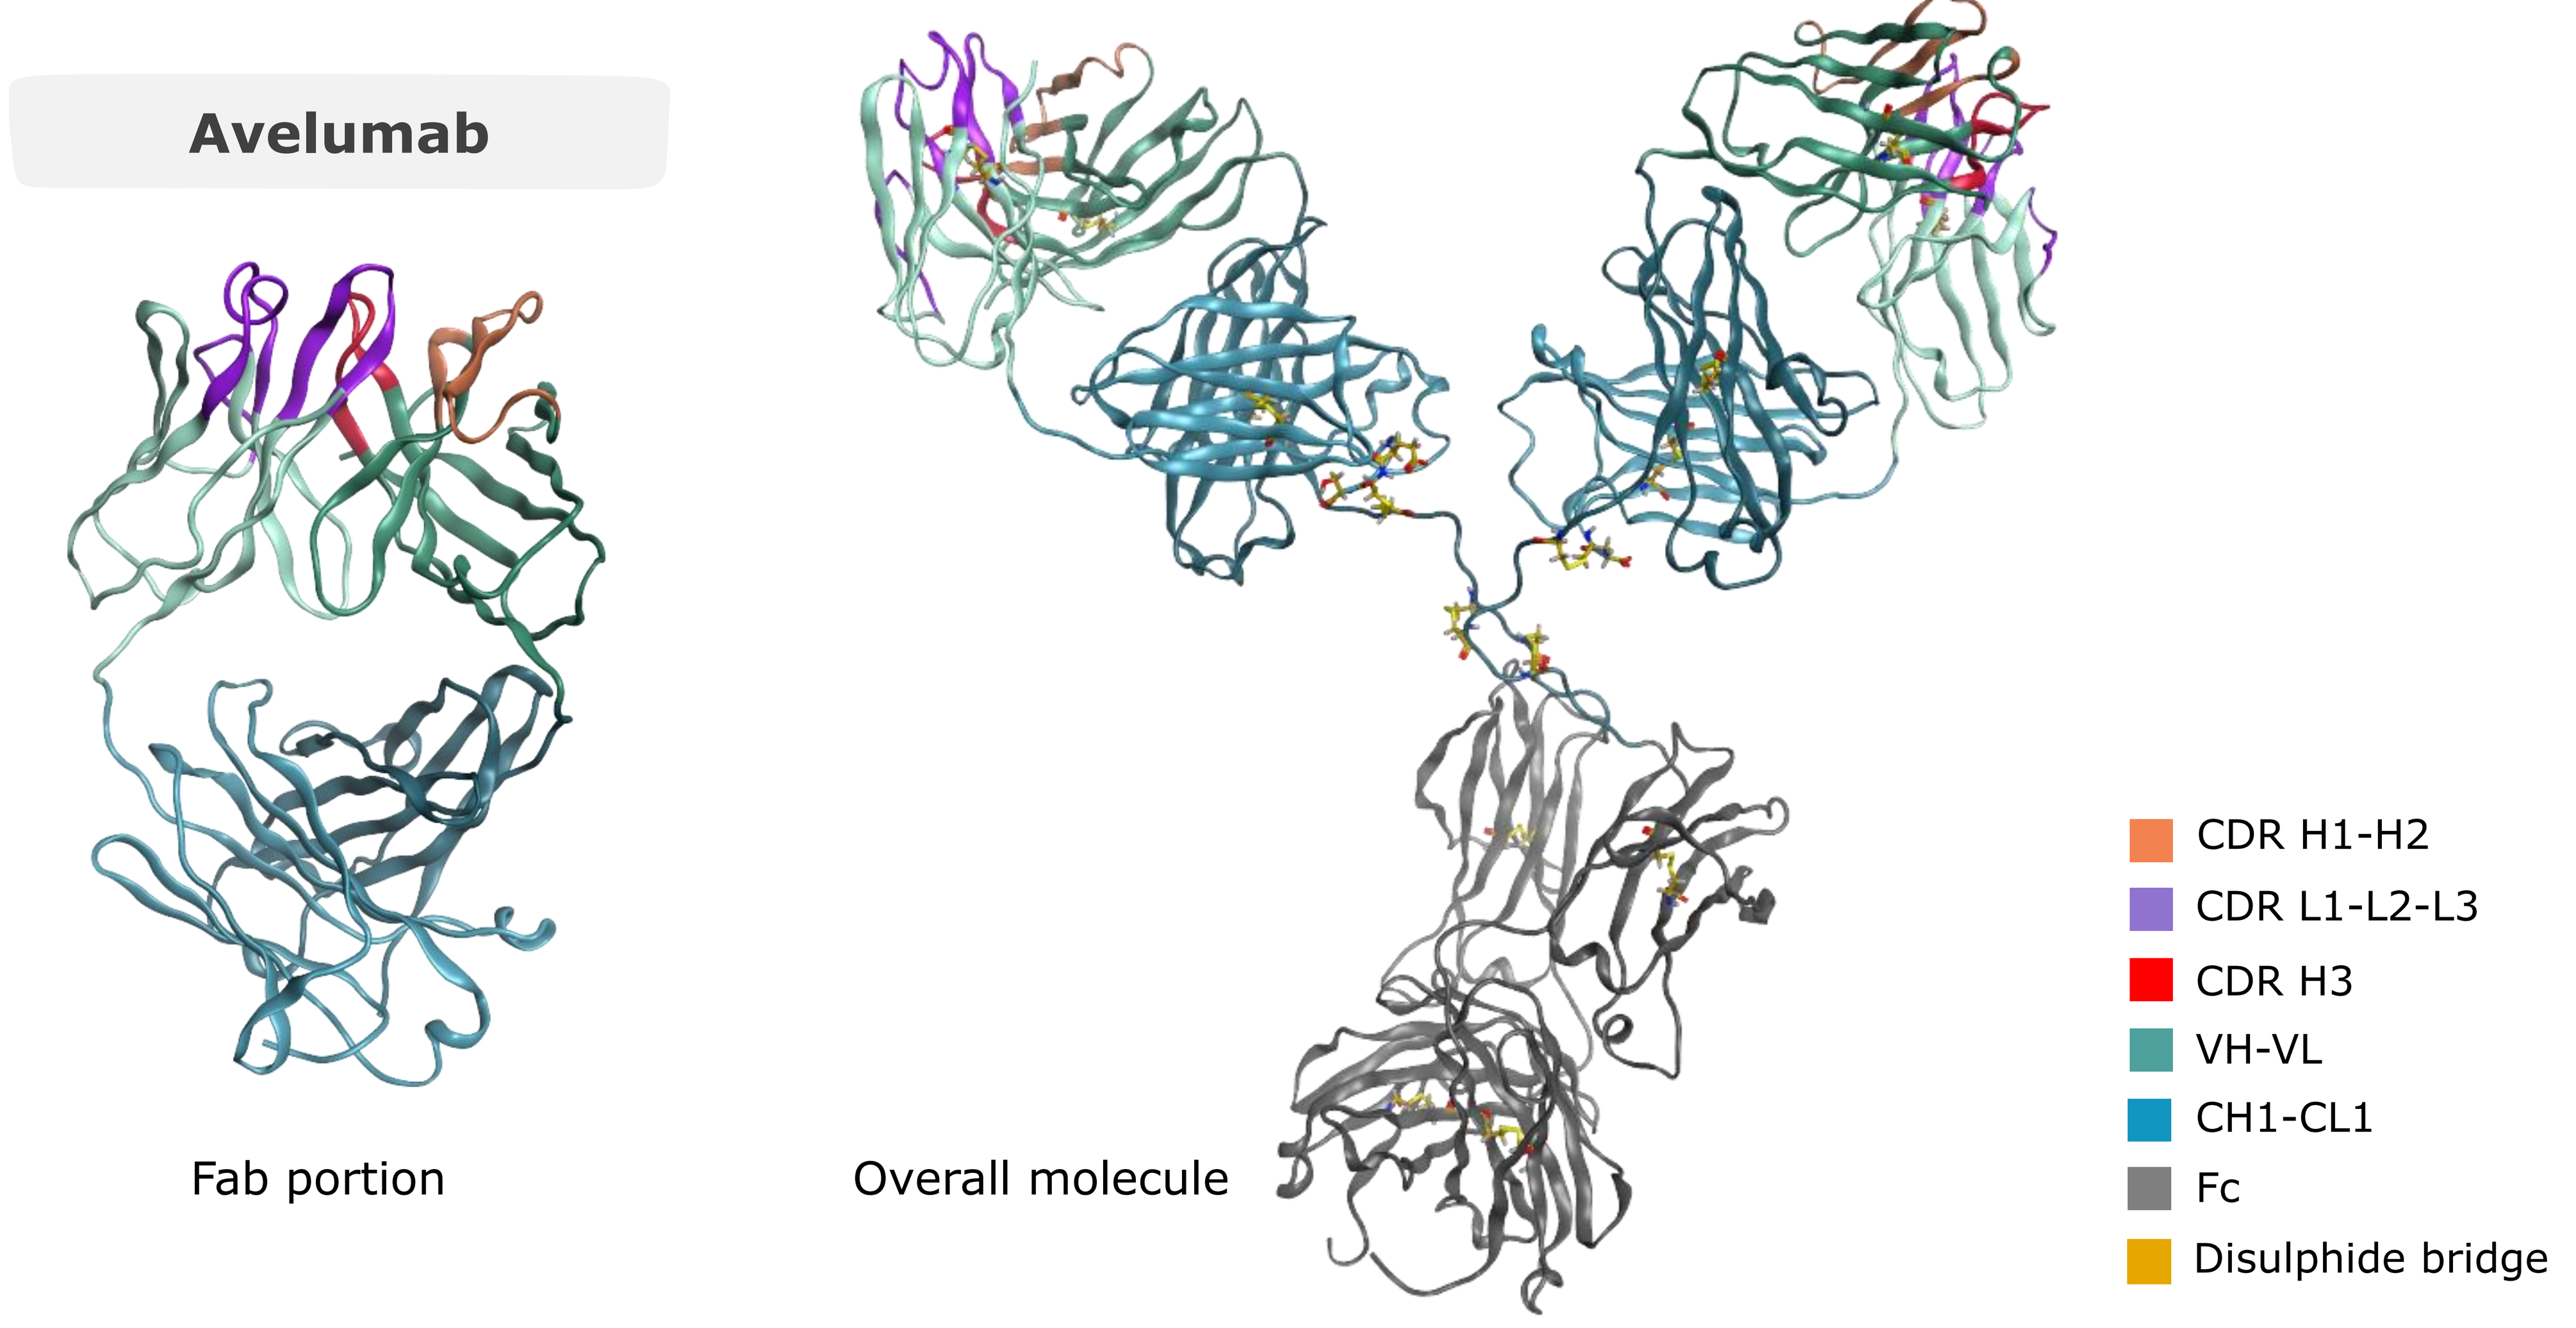
Avelumab
Fab portion
Overall molecule
CDR H1-H2
CDR L1-L2-L3
CDR H3
VH-VL
CH1-CL1
Fc
Disulphide bridge

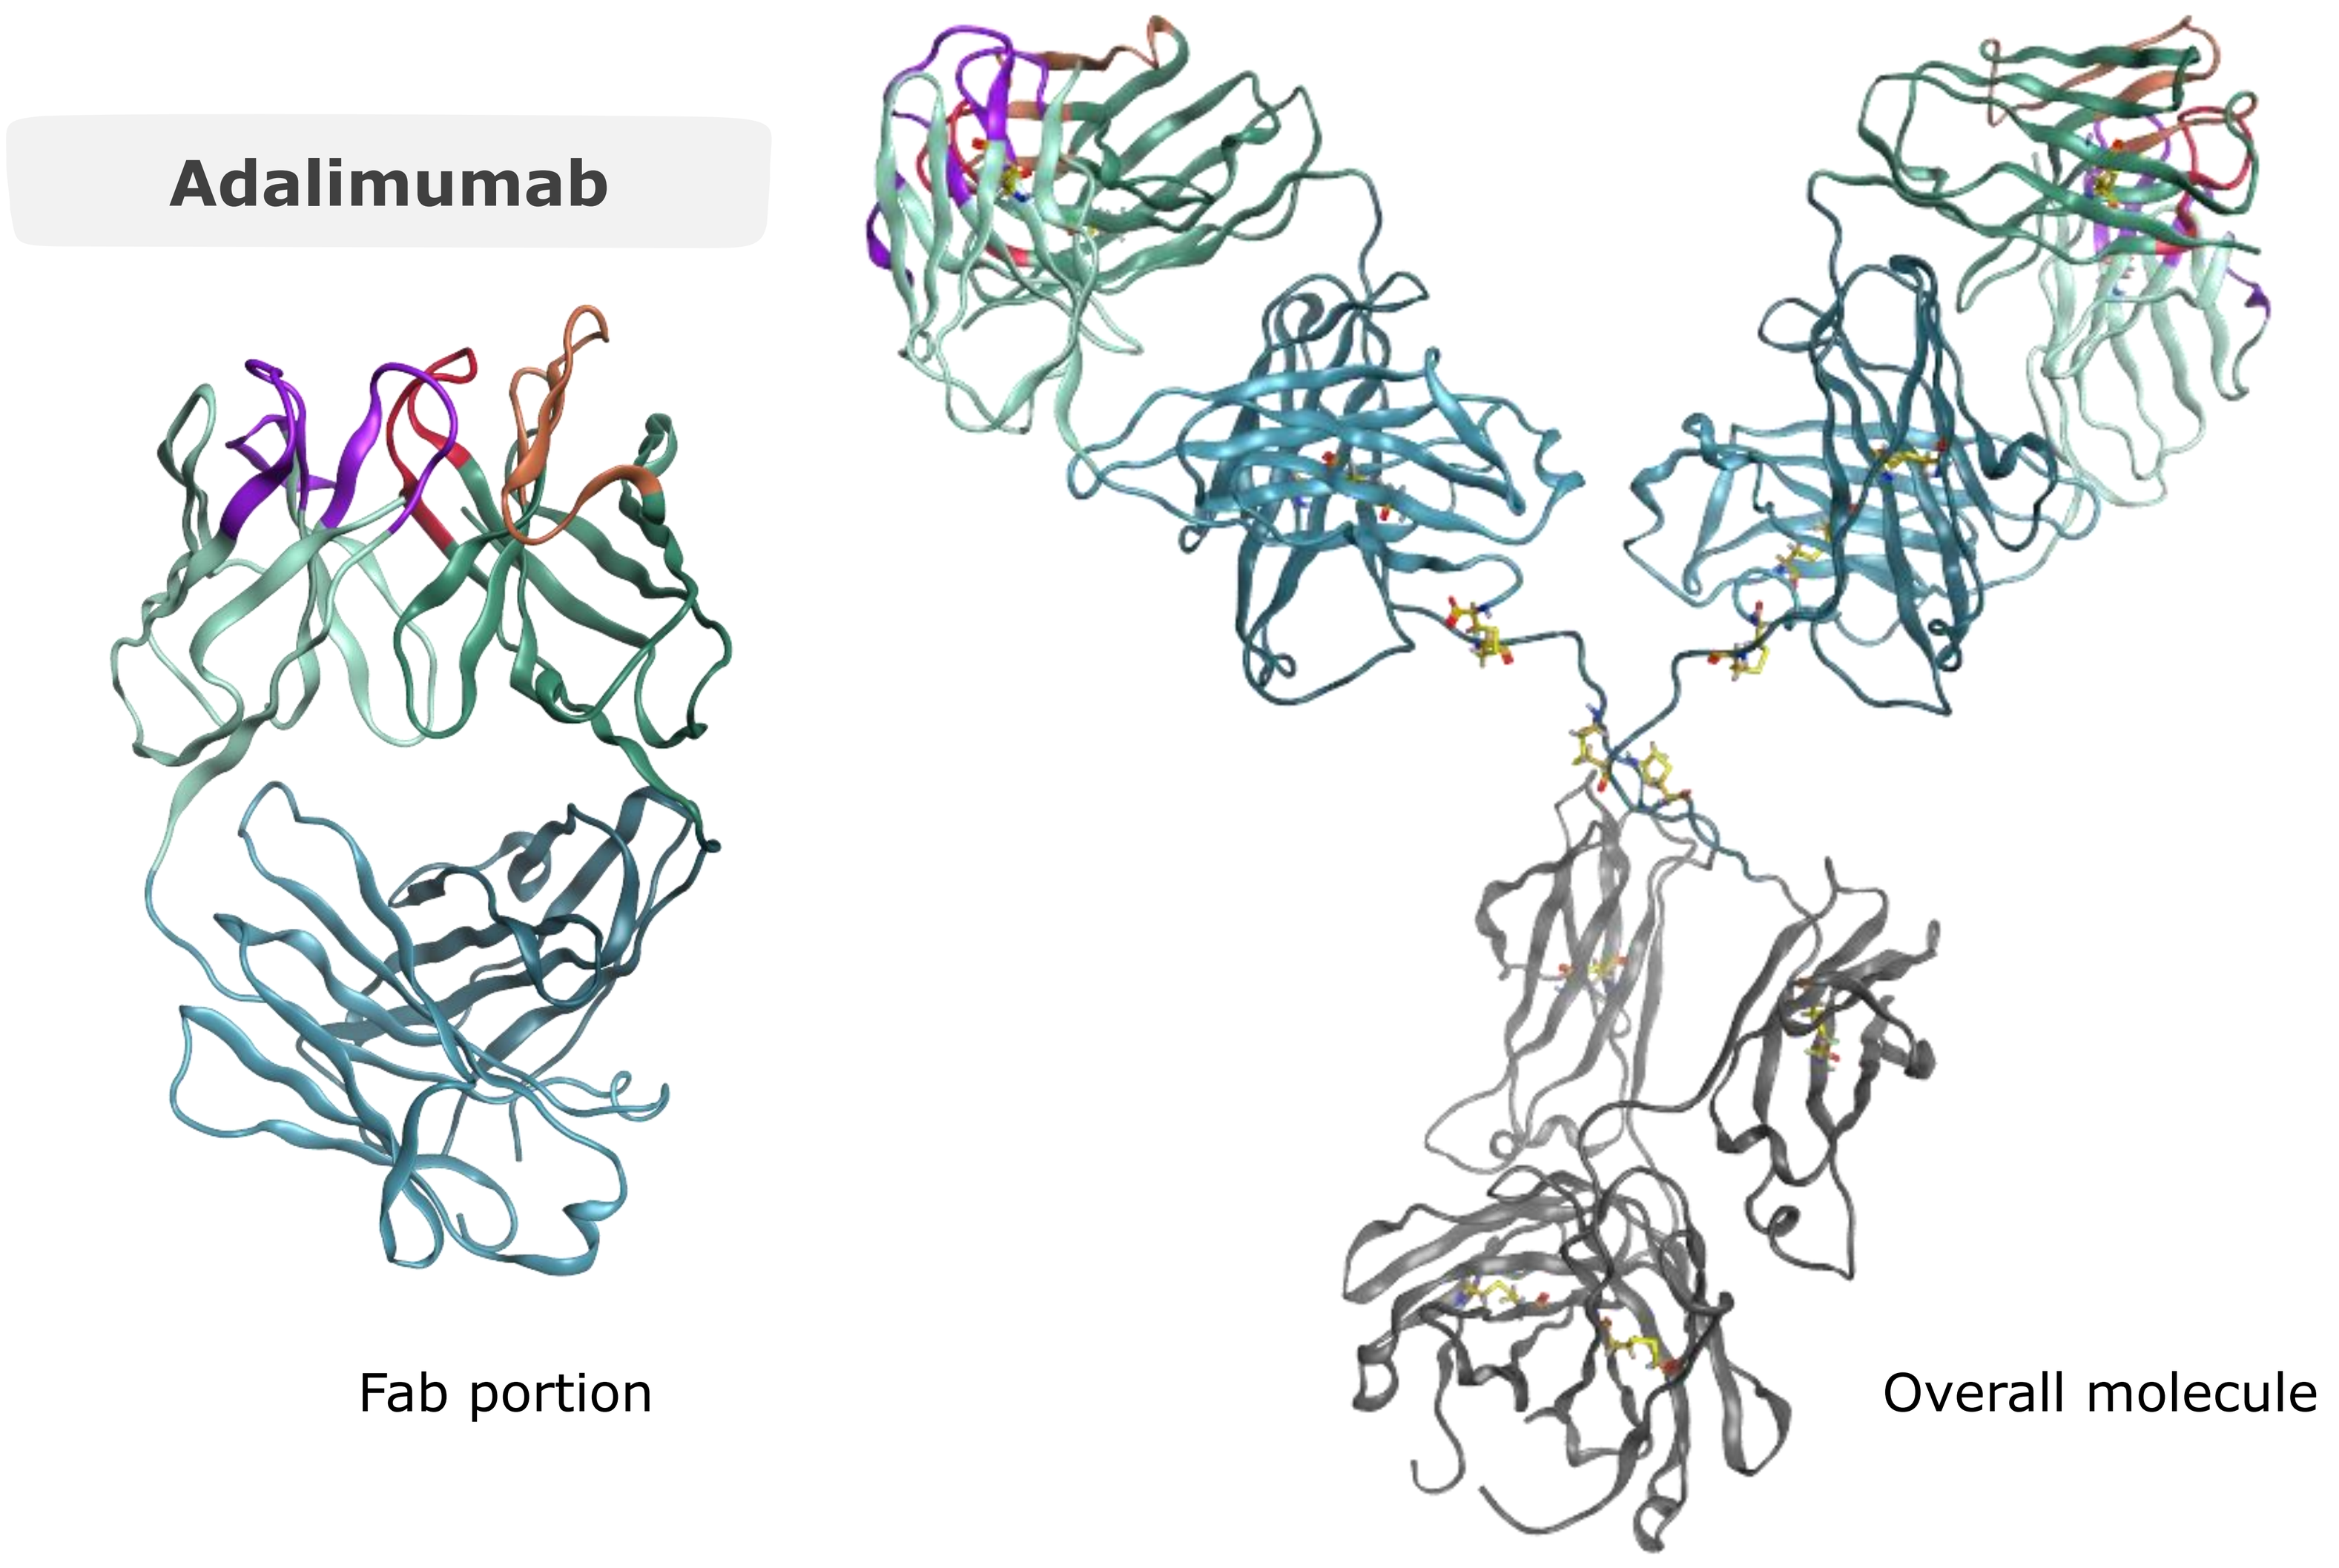
Adalimumab
Fab portion
Overall molecule

IgG1κ VS IgG1λ

The structural superposition of two models shows differences at CDRs, due to sequence heterogeneity, and a quite different orientation, mainly concentrated at the Fab C-terminal, close to hinge region. This feature may be influenced by the residue Ser216 displayed only in the λLC of avelumab (in green) and absent in adalimumab (in grey).

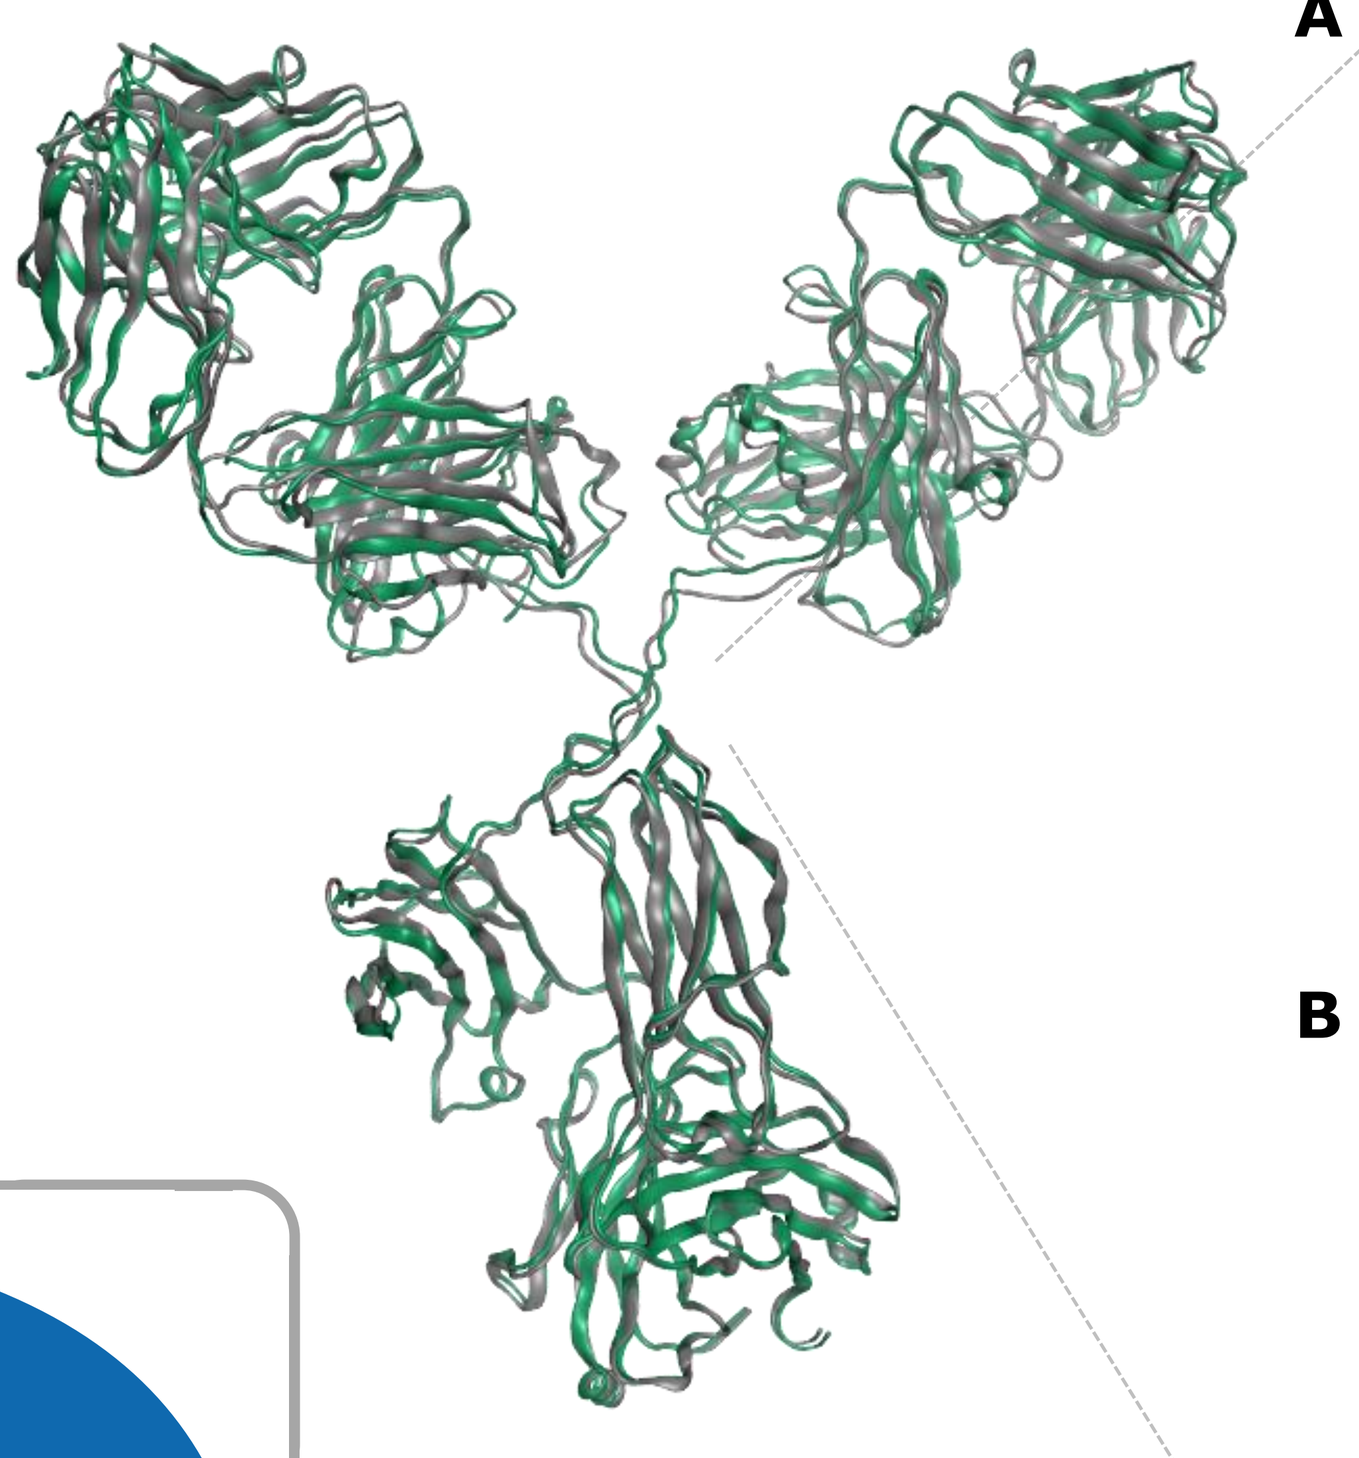

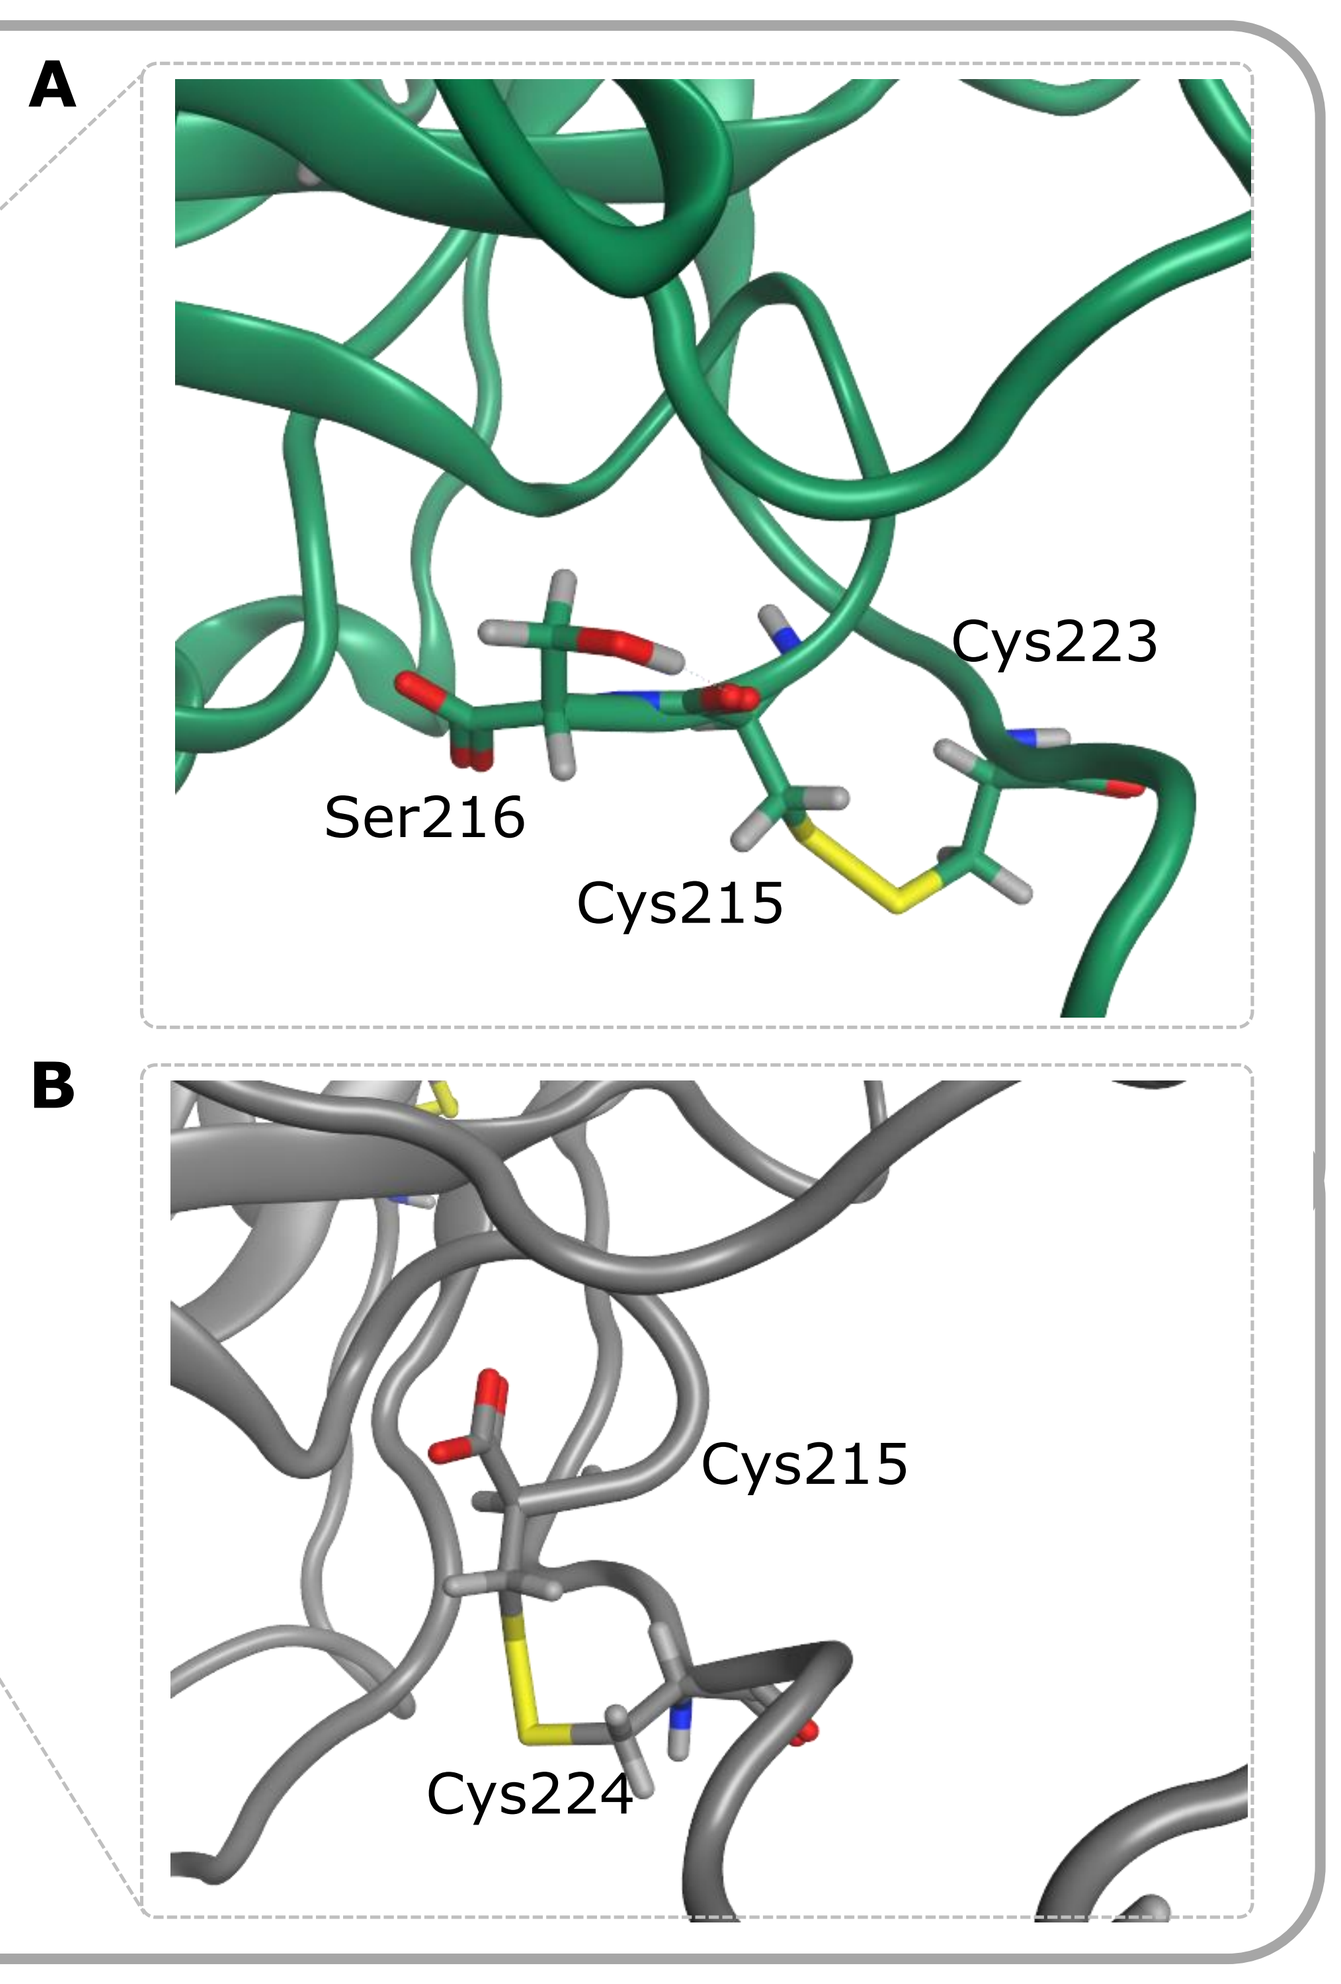
A
Cys223
Ser216
Cys215
B
Cys215
Cys224

(A) The avelumab hinge region with the interchain disuphide bridge. **(B)** The adalimumab hinge region.

CONCLUSIONS

- A chimeric approach is the most suitable to build homology models of mAbs, for both λ and κ light chains
- Ser216 of λ chains can induce slight differences in IgG1λ, expecially on the hinge region
- This structural difference could be responsible for different physical-chemical properties between IgG1 subtypes
- The structural approach proposed can be useful in CQA identification and assessment

References

1. *Food and Drug Administration CDER. Guidance for industry, Q8 pharmaceutical development (May 2006)*
2. *Janeway CA Jr, T. P. (2001); 5th edition. New York: Garland Science*
3. *Zhang et al, JBC, 2013; 288(23):16371-82*
4. *Toughiri R et al, MABS, 2016; 1276-1285*
5. *Merck-Pfizer Alliance. "Merck-Pfizer Alliance Avelumab Fact Sheet"(PDF). Retrieved 2 December 2015*
6. *Mease PJ, Therapeutics and Clinical Risk Management. 2007; 3(1):133-148*
7. *Hao G et al, J Mol Recognit., 2015; (4):269-76*
8. *Saphire EO et al, Science, 2001; 293(5532):1155-9*
9. *Molecular Operating Environment (MOE), 2013.08; Chemical Computing Group ULC, 2018*

Disclosures
Merck Serono, Guidonia Montecelio-Rome, Italy is an affiliate of Merck KGaA, Darmstadt, Germany.
Please note that avelumab has been approved in various countries for the treatment of metastatic Merkel cell carcinoma and in the US for treatment of advanced urothelial carcinoma progressed after platinum-containing treatment.